Real-life factors influencing journal selection

Nisha Sheikh[a,*], David Gothard[b], Anna Timbrell[b], Elise Blankenship[c]
[a]Ipsen, Abingdon, UK; [b]Oxford PharmaGenesis, Oxford, UK; [c]Ipsen, Cambridge, Massachusetts, USA
*At the time the survey was performed

14

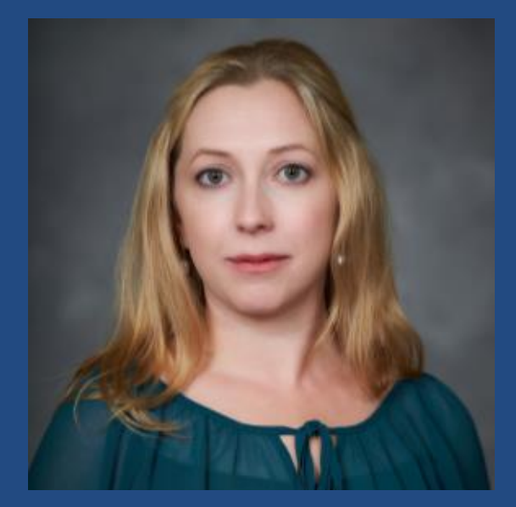

For further information, please send your question(s) to elise.blankenship@ipsen.com
For audio commentary, please click the icon.
To download the poster, please click the icon.

Copies of this eposter are for personal use only and may not be reproduced without written permission from the authors.

Objective

- Criteria exist to guide author selection and engagement for manuscript development. However, how identification and selection of the target journal for submission are determined is a variable process.
- To understand the journal shortlisting process, across all therapy areas, including the key factors and individuals affecting choice, and gather insights from those involved in publishing research.

Research design and methods

- An 18-item, online survey was developed to gather information on individuals' experiences of selecting journals, including questions on choice responsibility, use of selection tools, and review timings and cost.
- Circulated via LinkedIn, Open Pharma, the International Society for Medical Publication Professionals (ISMPP) Forum, the Medical Affairs Professional Society (MAPS) app MAPS Connect, and personal email invitations, the survey was active during the period 8–25 November 2021.

Results

- In total, 62 respondents completed the survey, with the majority from North America (44%) and Western Europe (39%) (**Panel 1**).
- Three-quarters of respondents were from medical communications agencies (39%) and pharmaceutical companies (37%) (**Panel 1**).
- Perceived quality of the articles published in the journal, journal metrics, and geographical reach were ranked the most important selection criteria; cost and personal invitation were ranked the least important (**Panel 2**).
- Only seven respondents (11%) reported that journal familiarity would not affect their choice (**Panel 2**).
- Two-thirds of respondents (66%) were aware of and used journal selection tools (e.g. Elsevier JournalFinder; Sylogent) (**Panel 3**).
- Almost half of respondents (45%) considered journal choice at manuscript initiation and 87% reported that authors had the final choice of journal (**Panel 4**).
- Most respondents reported that review times (87%) and publication times (85%) affected journal choice (**Panel 5**).
 - Of the 62 respondents, 68% reported 3 months or less as a reasonable time between submission and acceptance of a manuscript.
 - In total, 73% of respondents considered a period of up to 1 or 3 months as a reasonable time between manuscript acceptance and article publication.
- Most respondents (74%) expected to pay publication fees of US$2,500–5,000 per article and would pay more for open access or rapid review (**Panel 5**).
- Almost all respondents (59/62) checked for predatory journals; of these, 81% checked the indexing status (**Panel 6**).

Conclusions

- Our findings show that individuals involved in scientific publications:
 - select journals primarily based on perceived journal merit (quality, impact factor, reach, and speed of publication)
 - are not dissuaded by publication fees and opt for open access and rapid publication when available.
- Gathering insights from a wider audience would help reduce any bias.

Panel 1. Demographics of responders

Primary role

- 39% Pharmaceutical company
- 37% Medical communications
- 5% Academic
- 5% Independent consultant
- 5% Publisher
- 3% CRO or regulatory body
- 6% Other

Geographical location

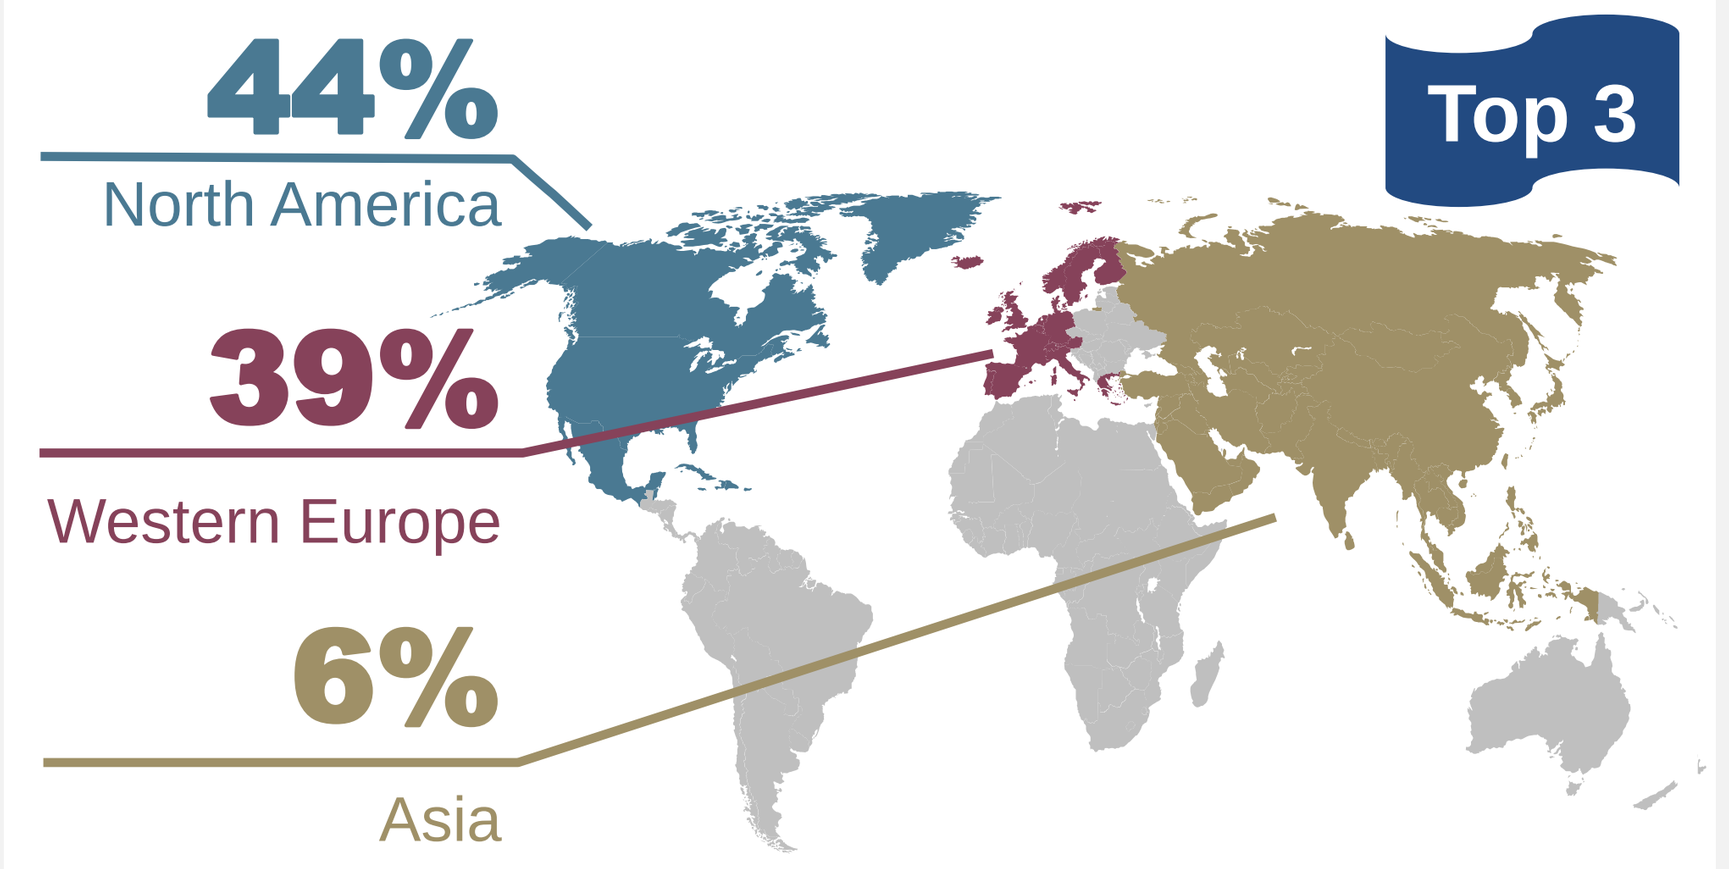
Top 3
44%
North America
39%
Western Europe
6%
Asia

Central/Eastern Europe, 5%; Australasia, 5%; Middle East, 2%.

Publication types that respondents have worked on

- 90% Clinical trial publications
- 68% Reviews or commentaries
- 58% Health economics
- 48% Preclinical research

Panel 2. Factors affecting journal selection

Journal selection criteria rankings

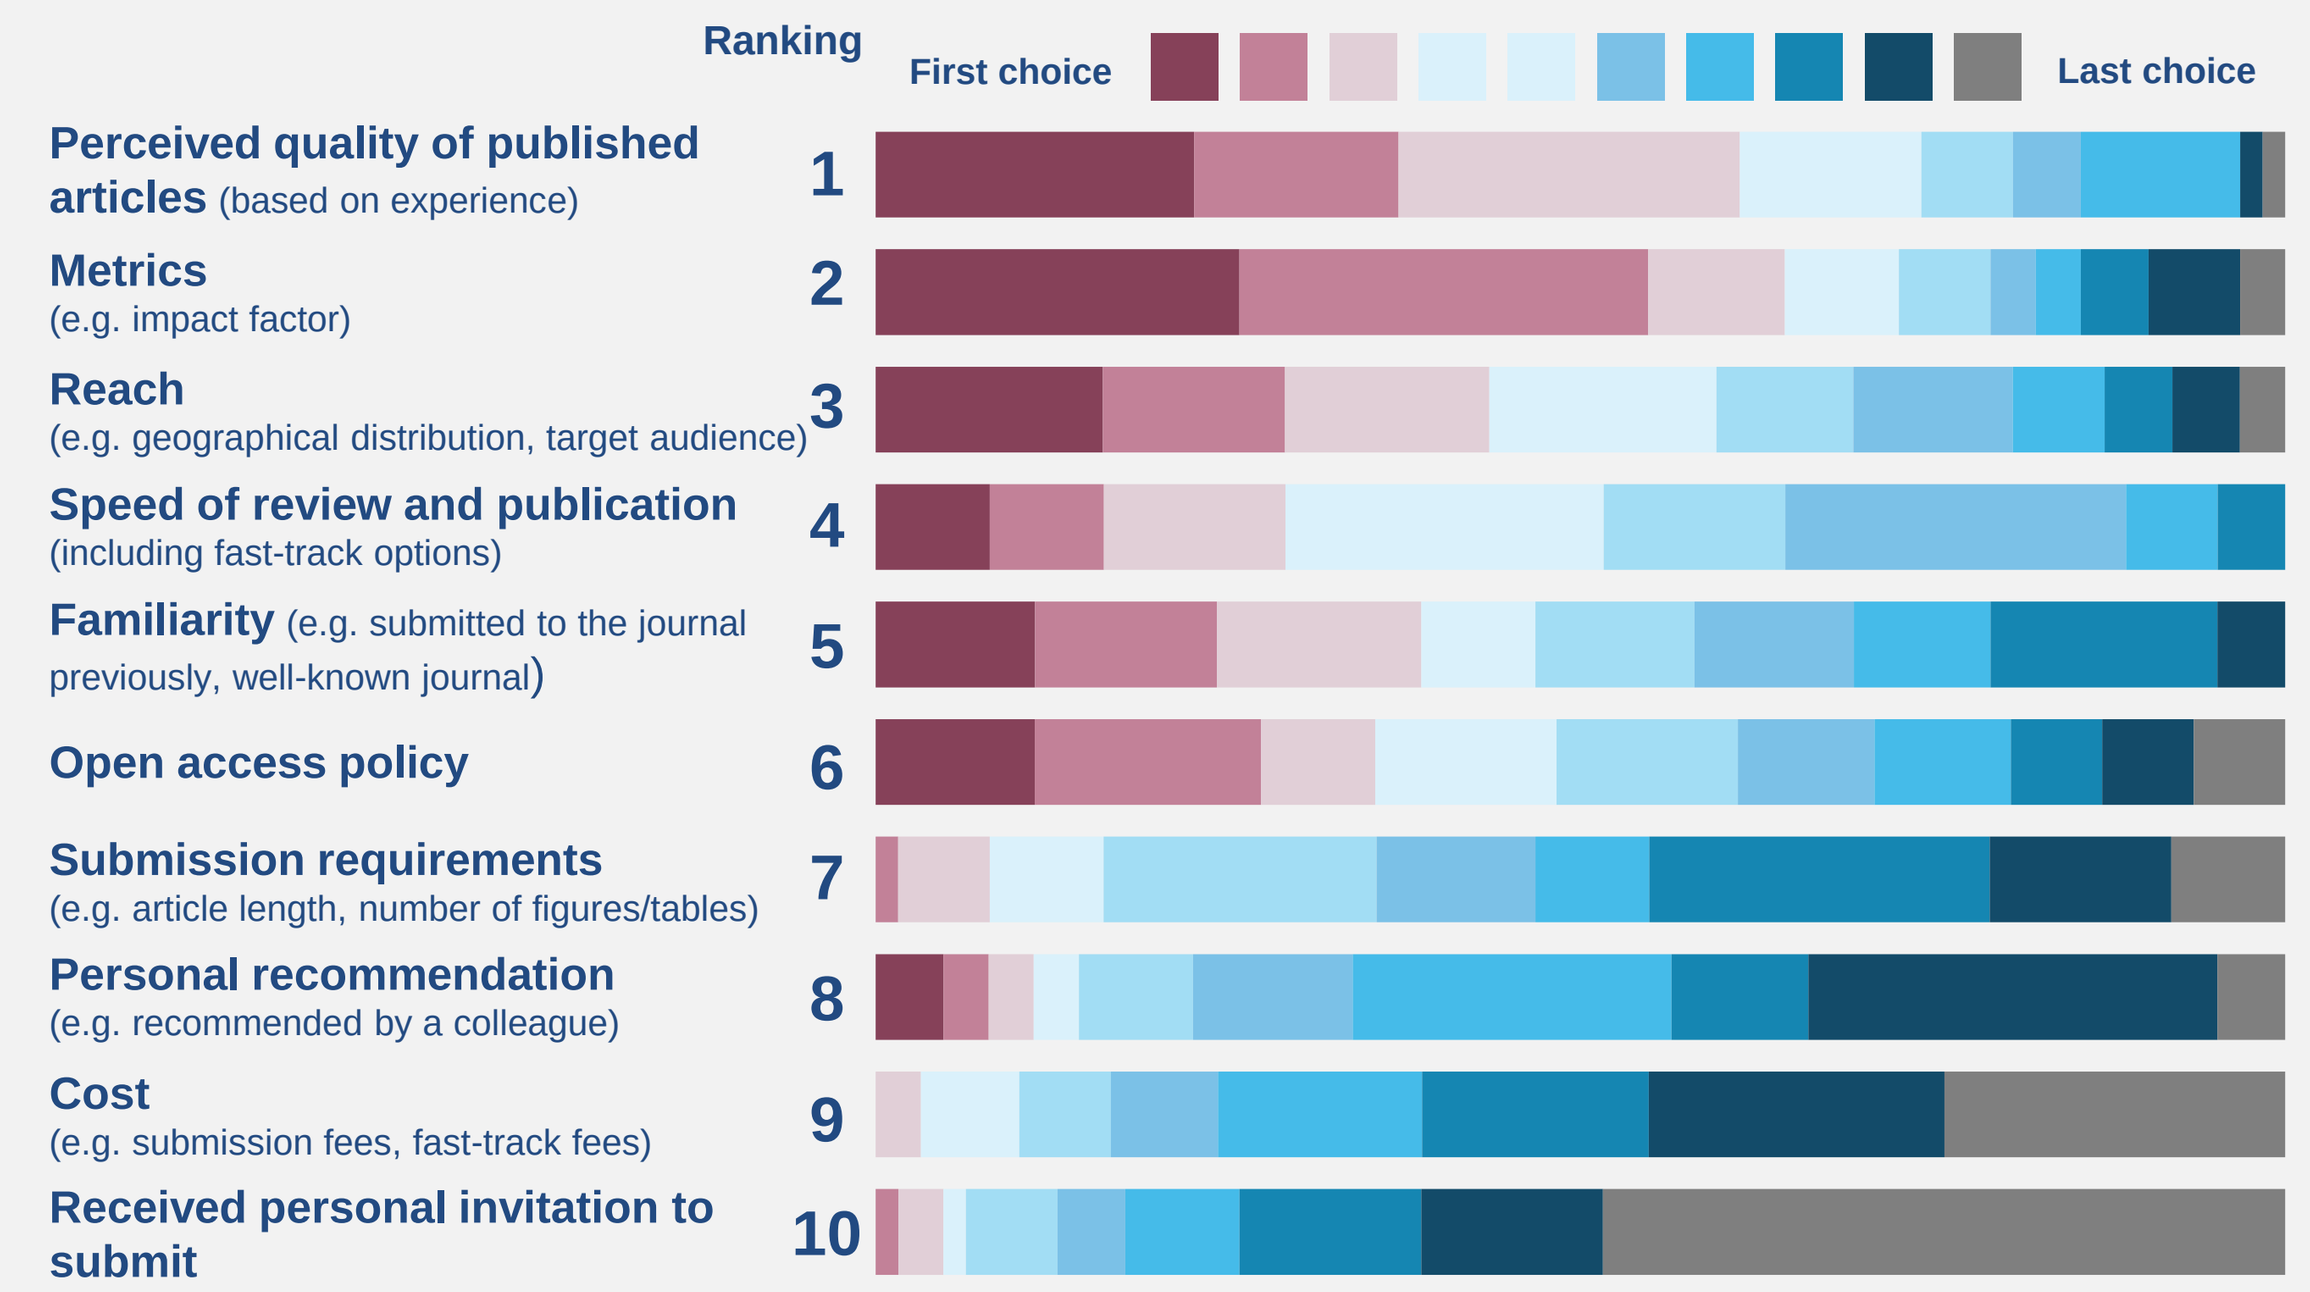
Ranking
First choice
Last choice
Perceived quality of published articles (based on experience) 1
Metrics (e.g. impact factor) 2
Reach (e.g. geographical distribution, target audience) 3
Speed of review and publication (including fast-track options) 4
Familiarity (e.g. submitted to the journal previously, well-known journal) 5
Open access policy 6
Submission requirements (e.g. article length, number of figures/tables) 7
Personal recommendation (e.g. recommended by a colleague) 8
Cost (e.g. submission fees, fast-track fees) 9
Received personal invitation to submit 10
Most important
Least important

Factors affecting journal choice

- 90% Journal is well-known
- 76% Previous experience with the journal
- 39% Personal recommendation
- 32% Journal editors known
- 89% Journal familiarity

Data represent the proportion of respondents who reported that the factor impacted their journal choice. Respondents could select more than one factor.

Panel 3. Journal selection tools

Aware of journal selection tools?

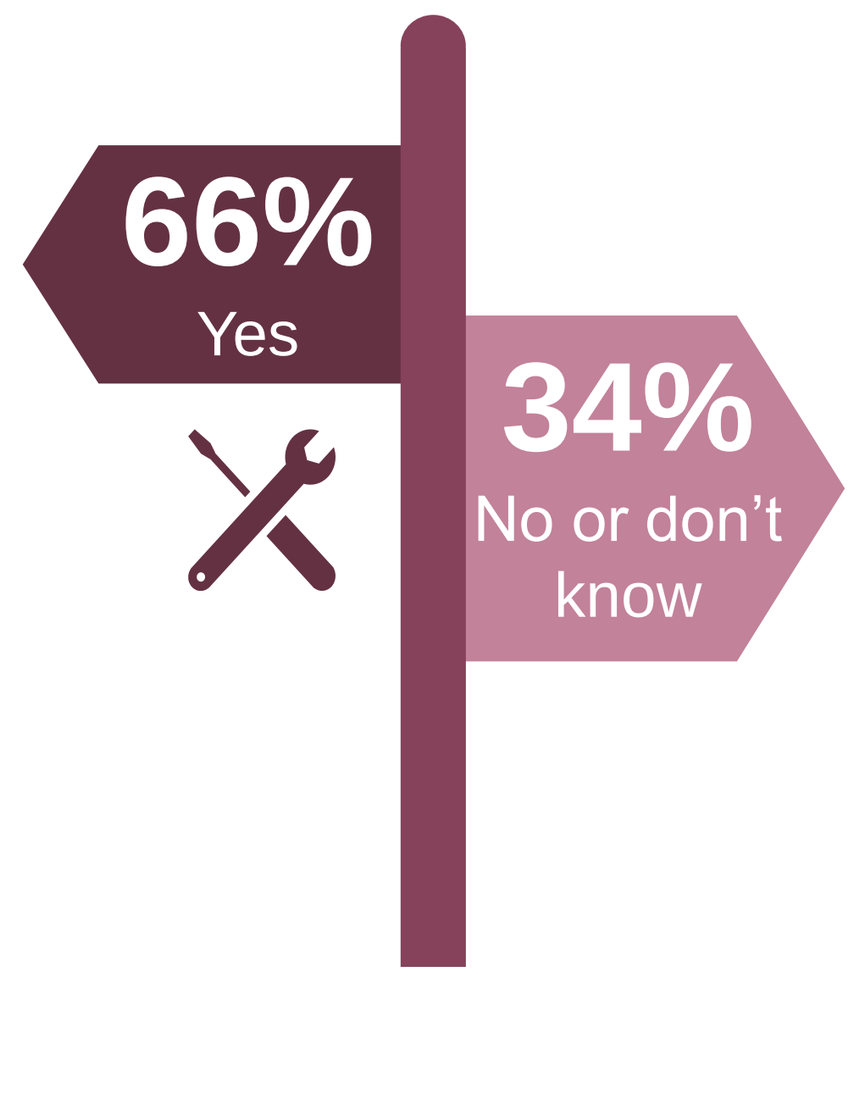
66%
Yes
34%
No or don't know

Familiarity with journal selection tools

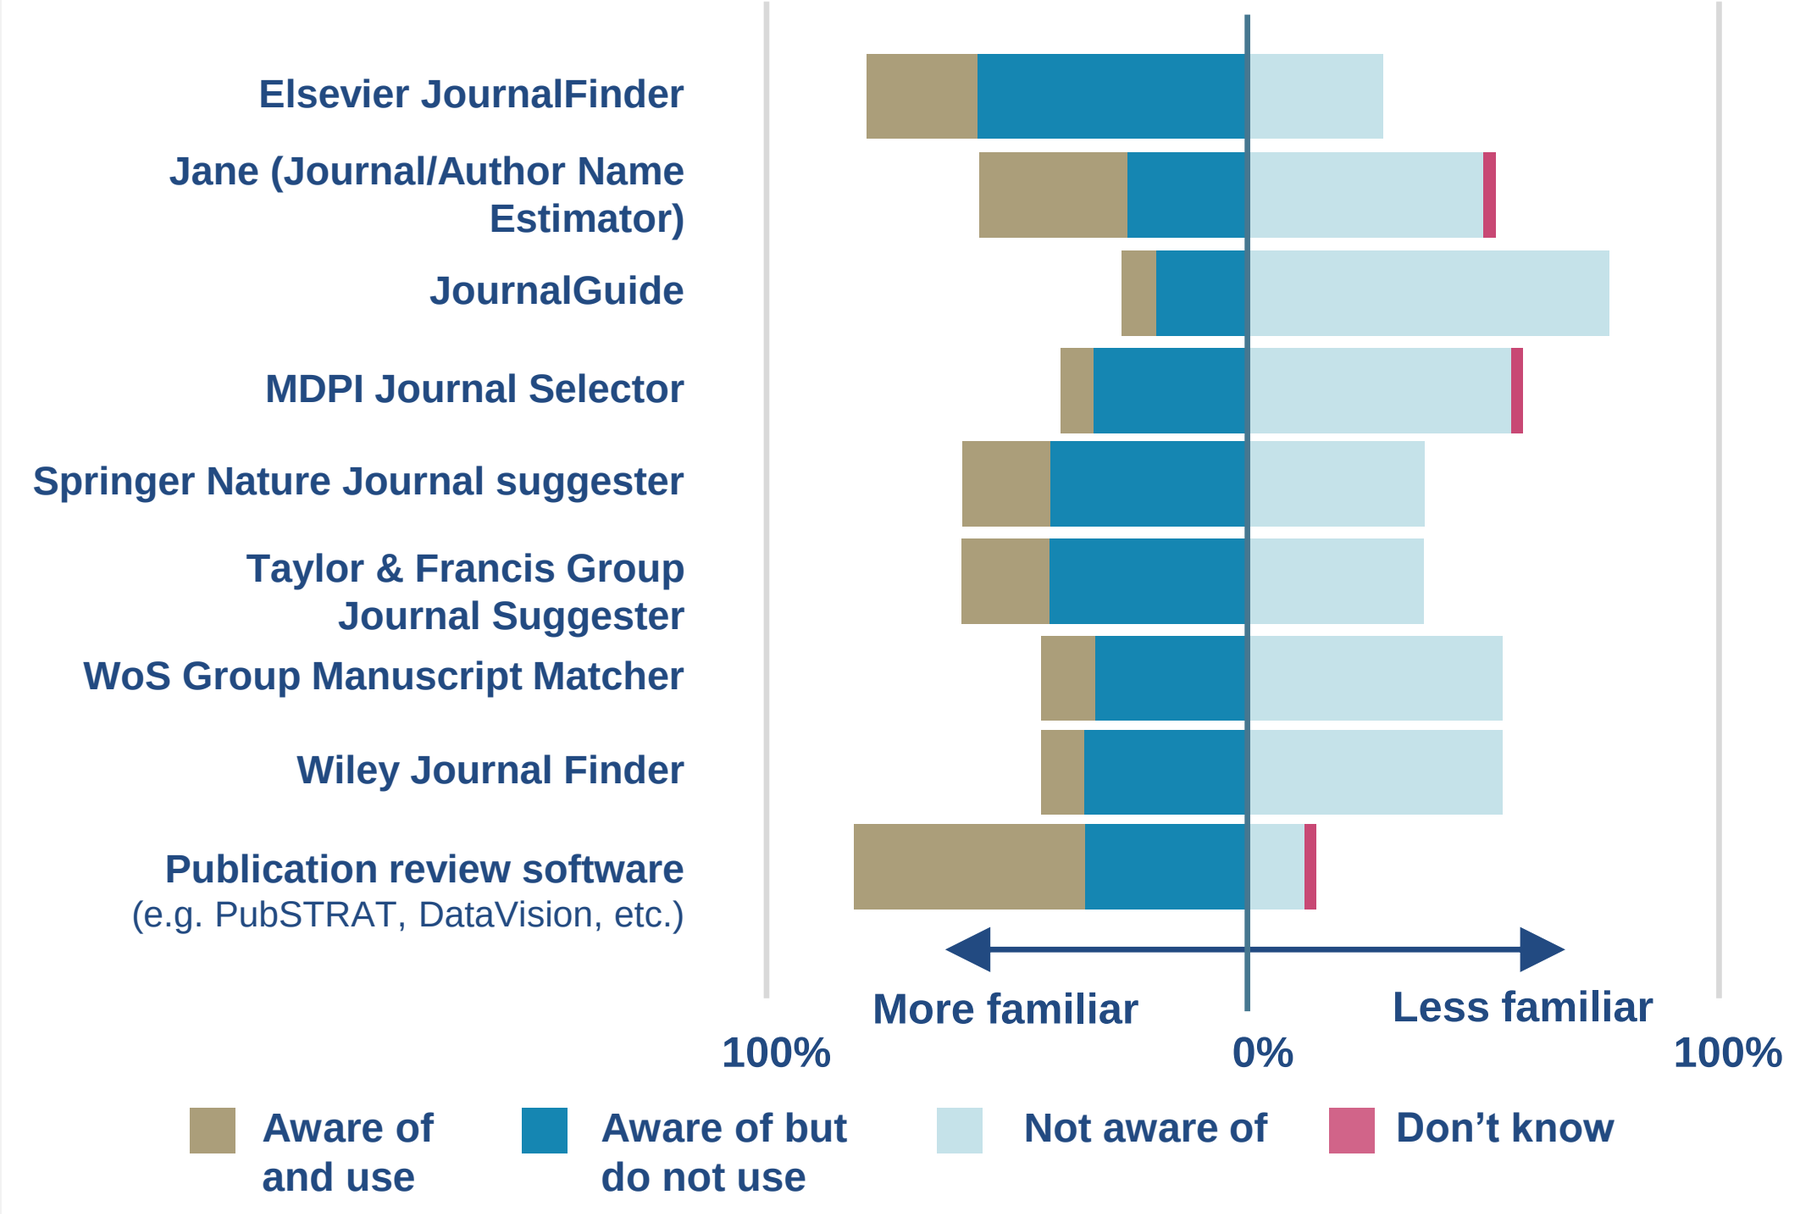
Elsevier JournalFinder
Jane (Journal/Author Name Estimator)
JournalGuide
MDPI Journal Selector
Springer Nature Journal suggester
Taylor & Francis Group Journal Suggester
WoS Group Manuscript Matcher
Wiley Journal Finder
Publication review software (e.g. PubSTRAT, DataVision, etc.)
More familiar
Less familiar
100%
0%
100%
Aware of and use
Aware of but do not use
Not aware of
Don't know

Percentage of respondent subpopulation (42/62) that answered 'Yes' or 'I don't know' when questioned on their awareness of selection tools.

Other journal selection tools noted by respondents[a]

Sponsor experience · Beall's list · PubsHub · Sylogent · Consultant · Internal team · Journals · Journal Selector · PubReMiner and MPIP · Medical writer · Experience library

[a]Journals and Sylogent were mentioned by two respondents. Other tools were mentioned by one respondent only.

Panel 4. When and who selects a journal

Stage at which respondents consider journal choice

45%	29%	15%	3%	2%
Start of manuscript development	Project initiation	Final data available	Initial data available	First draft of manuscript

'Don't know', 2%; 'Other', 5%.

Responsibility for journal selection

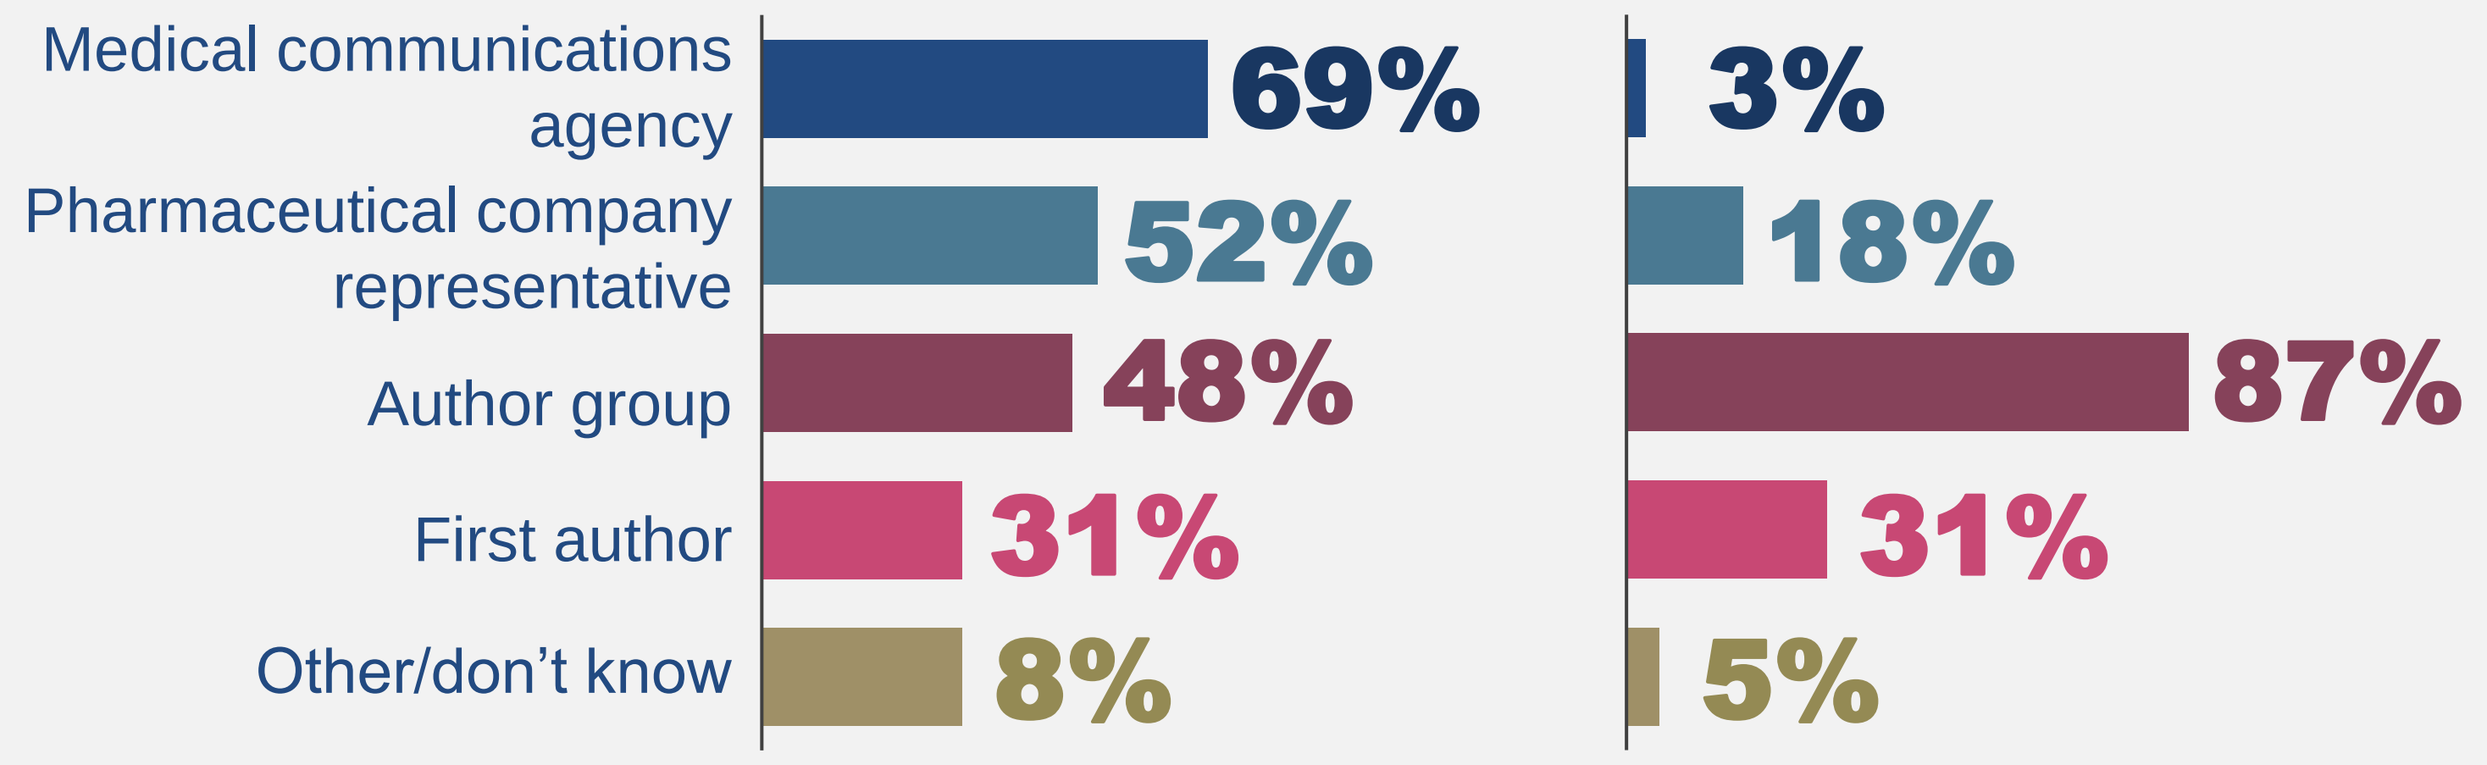
Initial journal list
Final journal choice
Medical communications agency 69% 3%
Pharmaceutical company representative 52% 18%
Author group 48% 87%
First author 31% 31%
Other/don't know 8% 5%

Panel 5. Publication costs and times

Costs expected by respondents for publication of a journal article

74% would expect to pay US$2,500–$5,000 per article

73% would pay more for **open access**

53% would pay more for **rapid review**

Perceptions of article process timings

Length of time considered reasonable for:

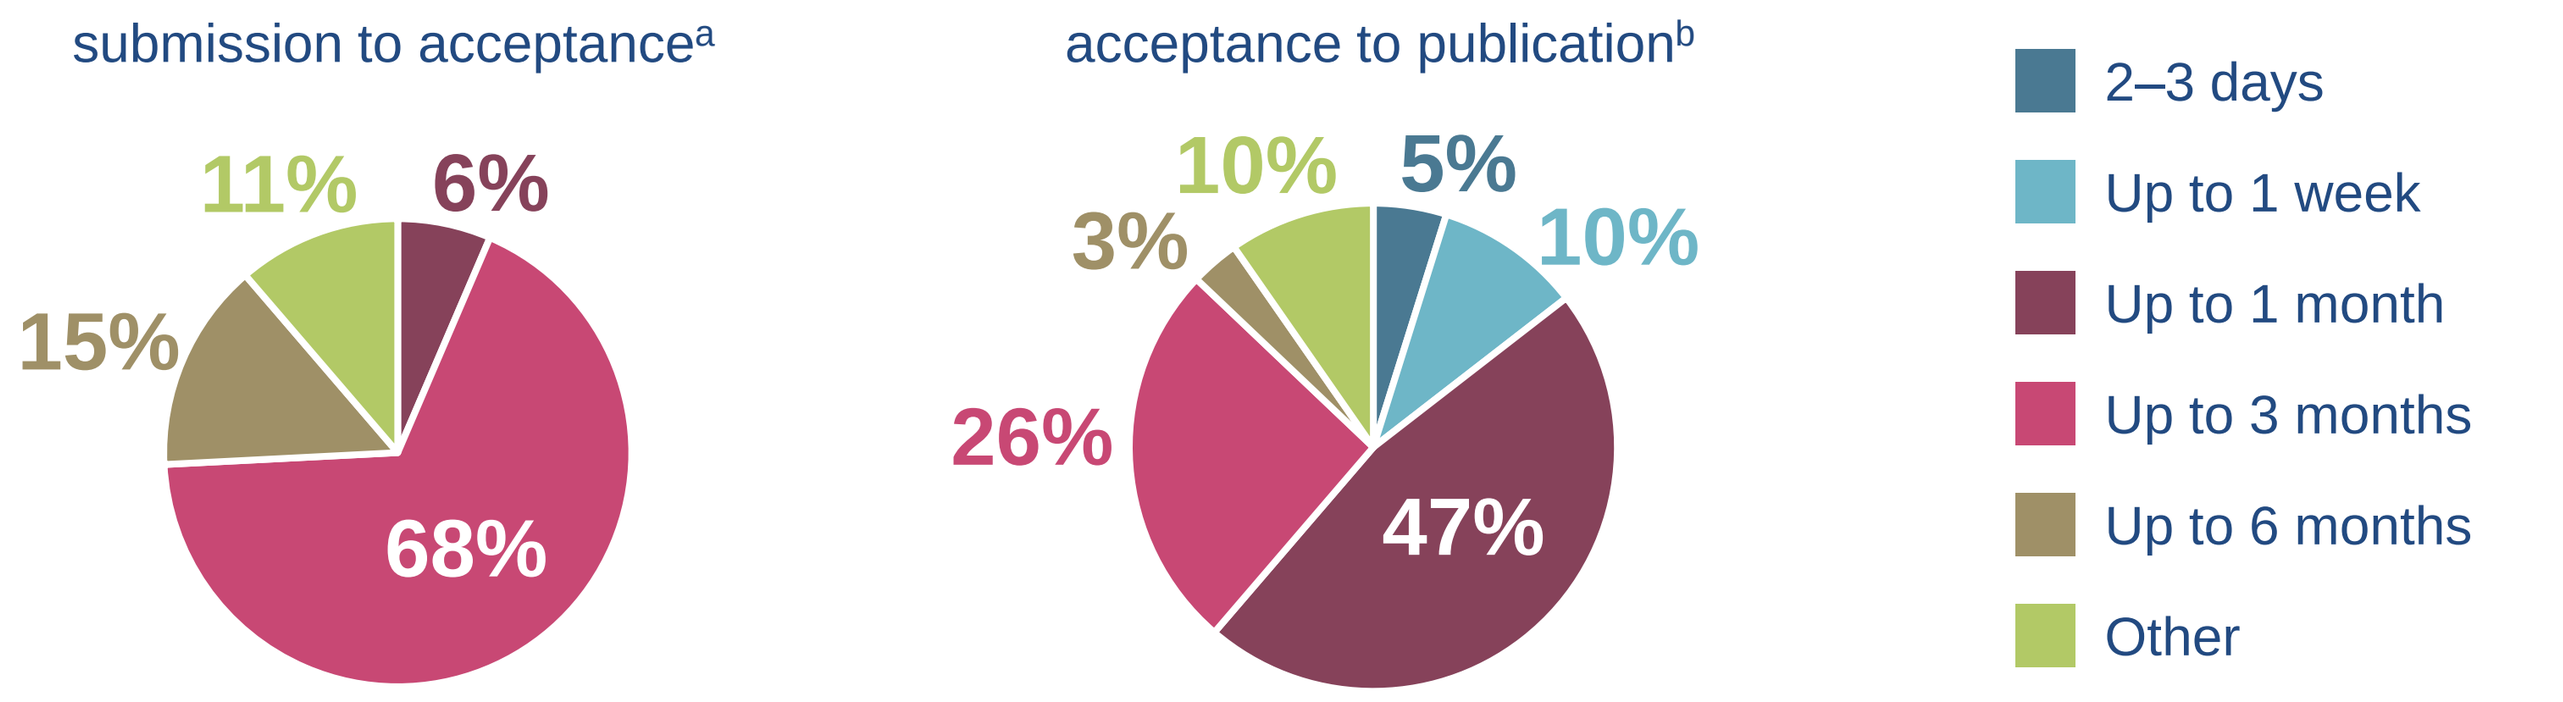
submission to acceptance[a]
11%
6%
15%
68%
acceptance to publication[b]
10%
5%
3%
10%
26%
47%
2–3 days
Up to 1 week
Up to 1 month
Up to 3 months
Up to 6 months
Other

[a]'Other' included 1–2 months (n = 1), < 2 months (n = 1), not more than 8–12 weeks (n =1), up to 4 months (n = 1), ~ 6 weeks (n = 1), and unspecified (n = 2).
[b]'Other' included < 2 weeks (n = 1), within 4 weeks (n = 1), < 2 months (n = 1), 16–17.2 weeks (n = 1), and unspecified (n = 2).

Panel 6. Predatory journals

Respondents who checked for predatory journals

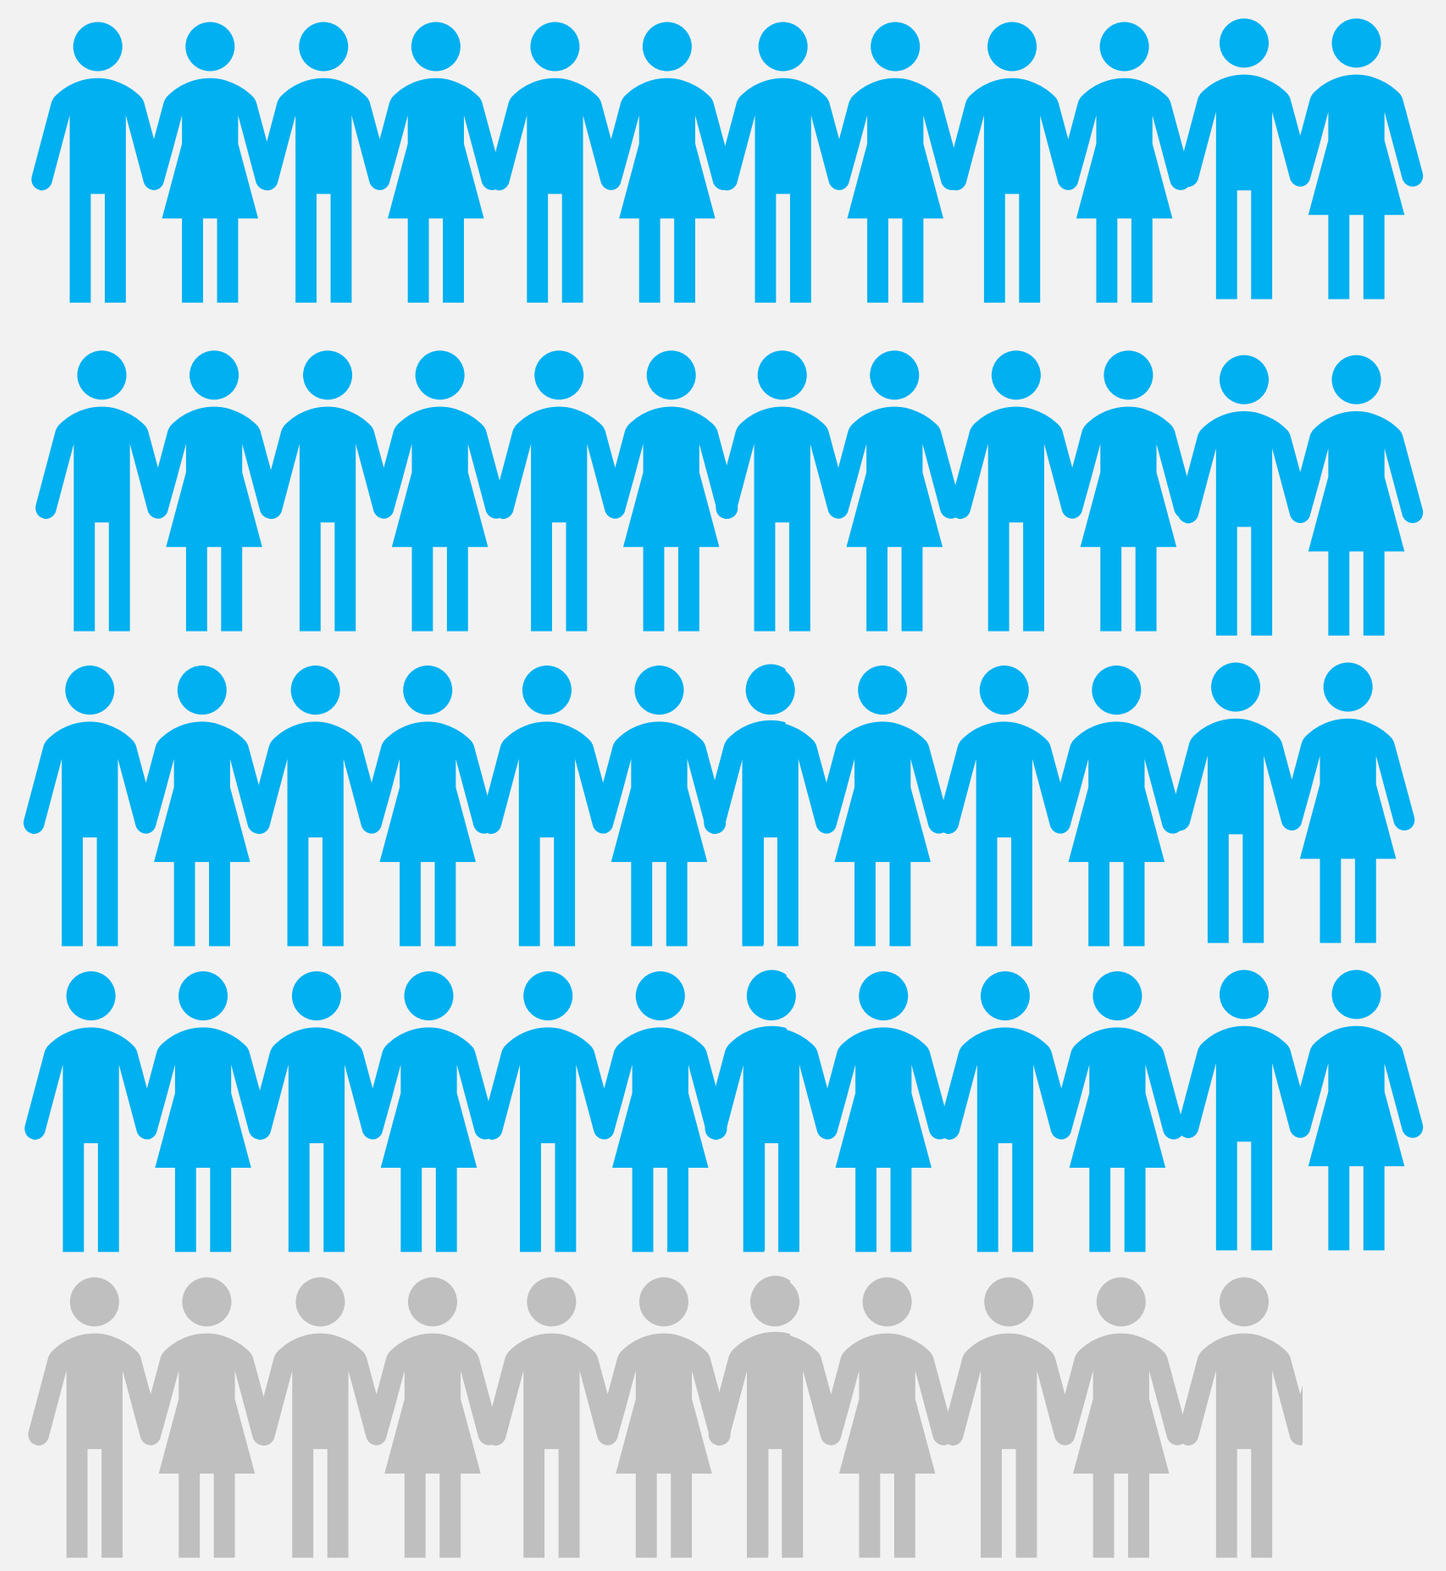

Of the 59 respondents who checked for predatory journals, **81%** checked the indexing status

Other approaches included:

- Beall's list (**51%**)
- journal has transparent/robust review process (**63%**)
- editorial board members (**39%**)
- standard checklists (**36%**)
- previous experience with journal (**36%**)

TAKE-HOME MESSAGE

In this survey of publication professionals, journal selection is driven by perceived journal merit more than cost, and the majority report that authors are responsible for the final journal choice.

Abbreviations CRO, clinical research organization; MPIP, Medical Publishing Insights & Practices; WoS, Web of Science.

Author Contributions All authors made substantial contributions to study conception/design, and acquisition/analysis/interpretation of data. All authors drafted the publication and revised it critically for important intellectual content and approved the final publication.

Disclosures NS: Employee of Ipsen at the time the survey was performed; DG and AT: Employees of Oxford PharmaGenesis; EB: Employee and shareholder of Ipsen.

Acknowledgments The authors thank all respondents who kindly completed the survey and thank Jo Gordon (PhD) of Oxford PharmaGenesis for providing writing, layout, and editorial support, which was sponsored by Ipsen in accordance with Good Publication Practice (GPP3) guidelines.

Presented at the 18th Annual Meeting of the International Society for Medical Publication Professionals (ISMPP), Washington, DC, USA | Virtual | 9–11 May 2022

This study was sponsored by Ipsen